Alteration of CD8+ CD45RC[int/neg] regulatory T cells functions in Multiple Sclerosis and correlates with disease severity

Naïl Benallegue, Bryan Nicol, Séverine Bézie, Hadrien Regue, Nadège Vimont, Léa Flippe, Alexandra Garcia, David A. Laplaud, Carole Guillonneau

▶ **To cite this version:**

Naïl Benallegue, Bryan Nicol, Séverine Bézie, Hadrien Regue, Nadège Vimont, et al.. Alteration of CD8+ CD45RC[int/neg] regulatory T cells functions in Multiple Sclerosis and correlates with disease severity. ECTRIMS 2018 (34th congress of the european committee for treatment and research in Multiple Sclerosis), Oct 2018, Berlin, Germany. inserm-02159755

HAL Id: inserm-02159755

https://inserm.hal.science/inserm-02159755

Submitted on 19 Jun 2019

HAL is a multi-disciplinary open access archive for the deposit and dissemination of scientific research documents, whether they are published or not. The documents may come from teaching and research institutions in France or abroad, or from public or private research centers.

L'archive ouverte pluridisciplinaire **HAL**, est destinée au dépôt et à la diffusion de documents scientifiques de niveau recherche, publiés ou non, émanant des établissements d'enseignement et de recherche français ou étrangers, des laboratoires publics ou privés.

Alteration of CD8+CD45RC^int/neg regulatory T cells functions in Multiple Sclerosis and correlates with disease severity

Naïl Benallegue[1,2,3,4], **Bryan Nicol**[1,2,3,4], **Séverine Bézie**[1,2,3], **Hadrien Regue**[1,2,3], **Nadège Vimont**[1,2,3], **Léa Flippe**[1,2,3], **Alexandra Garcia**[1,2,3], **David A. Laplaud**[1,2,3,4] **and Carole Guillonneau**[1,2,3,4].

[1]Center for Research in Transplantation and Immunology, INSERM, Université de Nantes, Nantes, France; [2]Institut de Transplantation Urologie Néphrologie (ITUN), CHU Nantes, Nantes, France; [3]LabEx IGO "Immunotherapy, Graft, Oncology", Nantes, France; [4]co-authors

Background

Autoimmune diseases can develop following pathological activation of autoreactive effector cells and/or, alternatively, after weakening of self-protective regulatory mechanisms. Most of the studies have focused on CD4$^+$ Tregs and the **role of CD8$^+$ Tregs in Multiple Sclerosis (MS) remains largely unexplored**. We previously reported the suppressive properties of rat and human **CD8$^+$CD45RC$^{int/neg}$ Treg cells**, expressing Foxp3 and acting through IFNg, TGFb and IL34 cytokines (Guillonneau, JCI, 2007; Bézie, JCI, 2015, Bézie, Front. Immunol., 2018). Thus, their **potency of suppression** make them strong candidates of disruptive immune tolerance, especially in MS where CD8$^+$ T cells play a major role. **Thus, the overreaching goal of this study is to define the role of CD8$^+$ regulatory T cell in MS pathogenesis**

Material & Methods

56 untreated relapsing-remitting MS patients and 52 age- and gender-matched healthy volunteers (HV) were recruited.

- Tregs were defined as CD3$^+$CD8$^+$ (or CD4$^-$) CD161^{low}Vα7$^-$CD45RC$^{int/neg}$ T cells. MAIT cells were excluded from all analysis.
- Staining: T cells were stimulated or not 5h with PMA+ionomycin including 4h with BFA before FACS depending on the marker investigated.
- Transcriptomic analysis: 3'-Digital Gene Expression Sequencing was performed on unstimulated cells. Analysis was performed using R packages and KEGG, Reactome and Gene Ontology databases for Gene Set Enrichment Analysis (GSEA).

CD8+CD45RC^INT Tregs frequency in blood is reduced during exacerbation whereas CD8+CD45RC^NEG Tregs are more frequent with enhanced suppressive functions

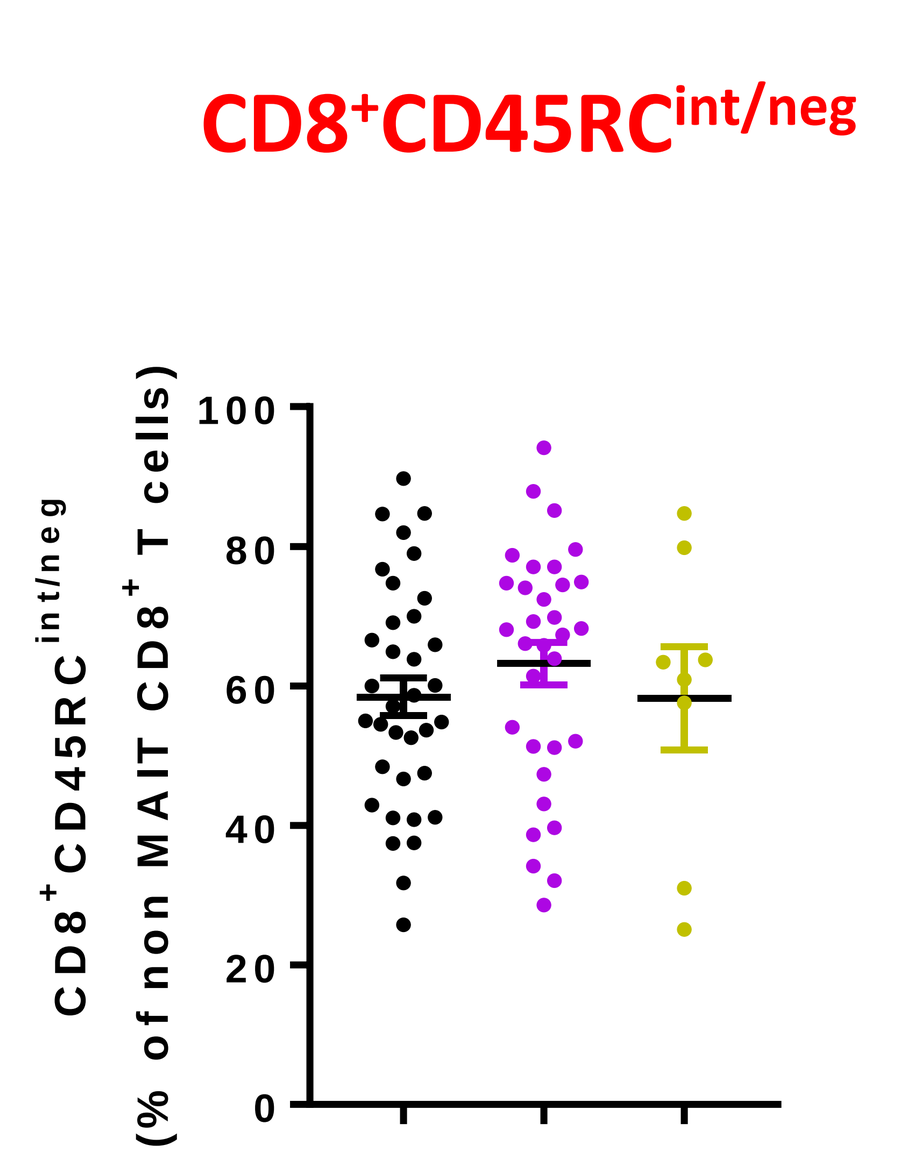
CD8+CD45RC int/neg
CD8+CD45RC int/neg (% of non MAIT CD8+ T cells)

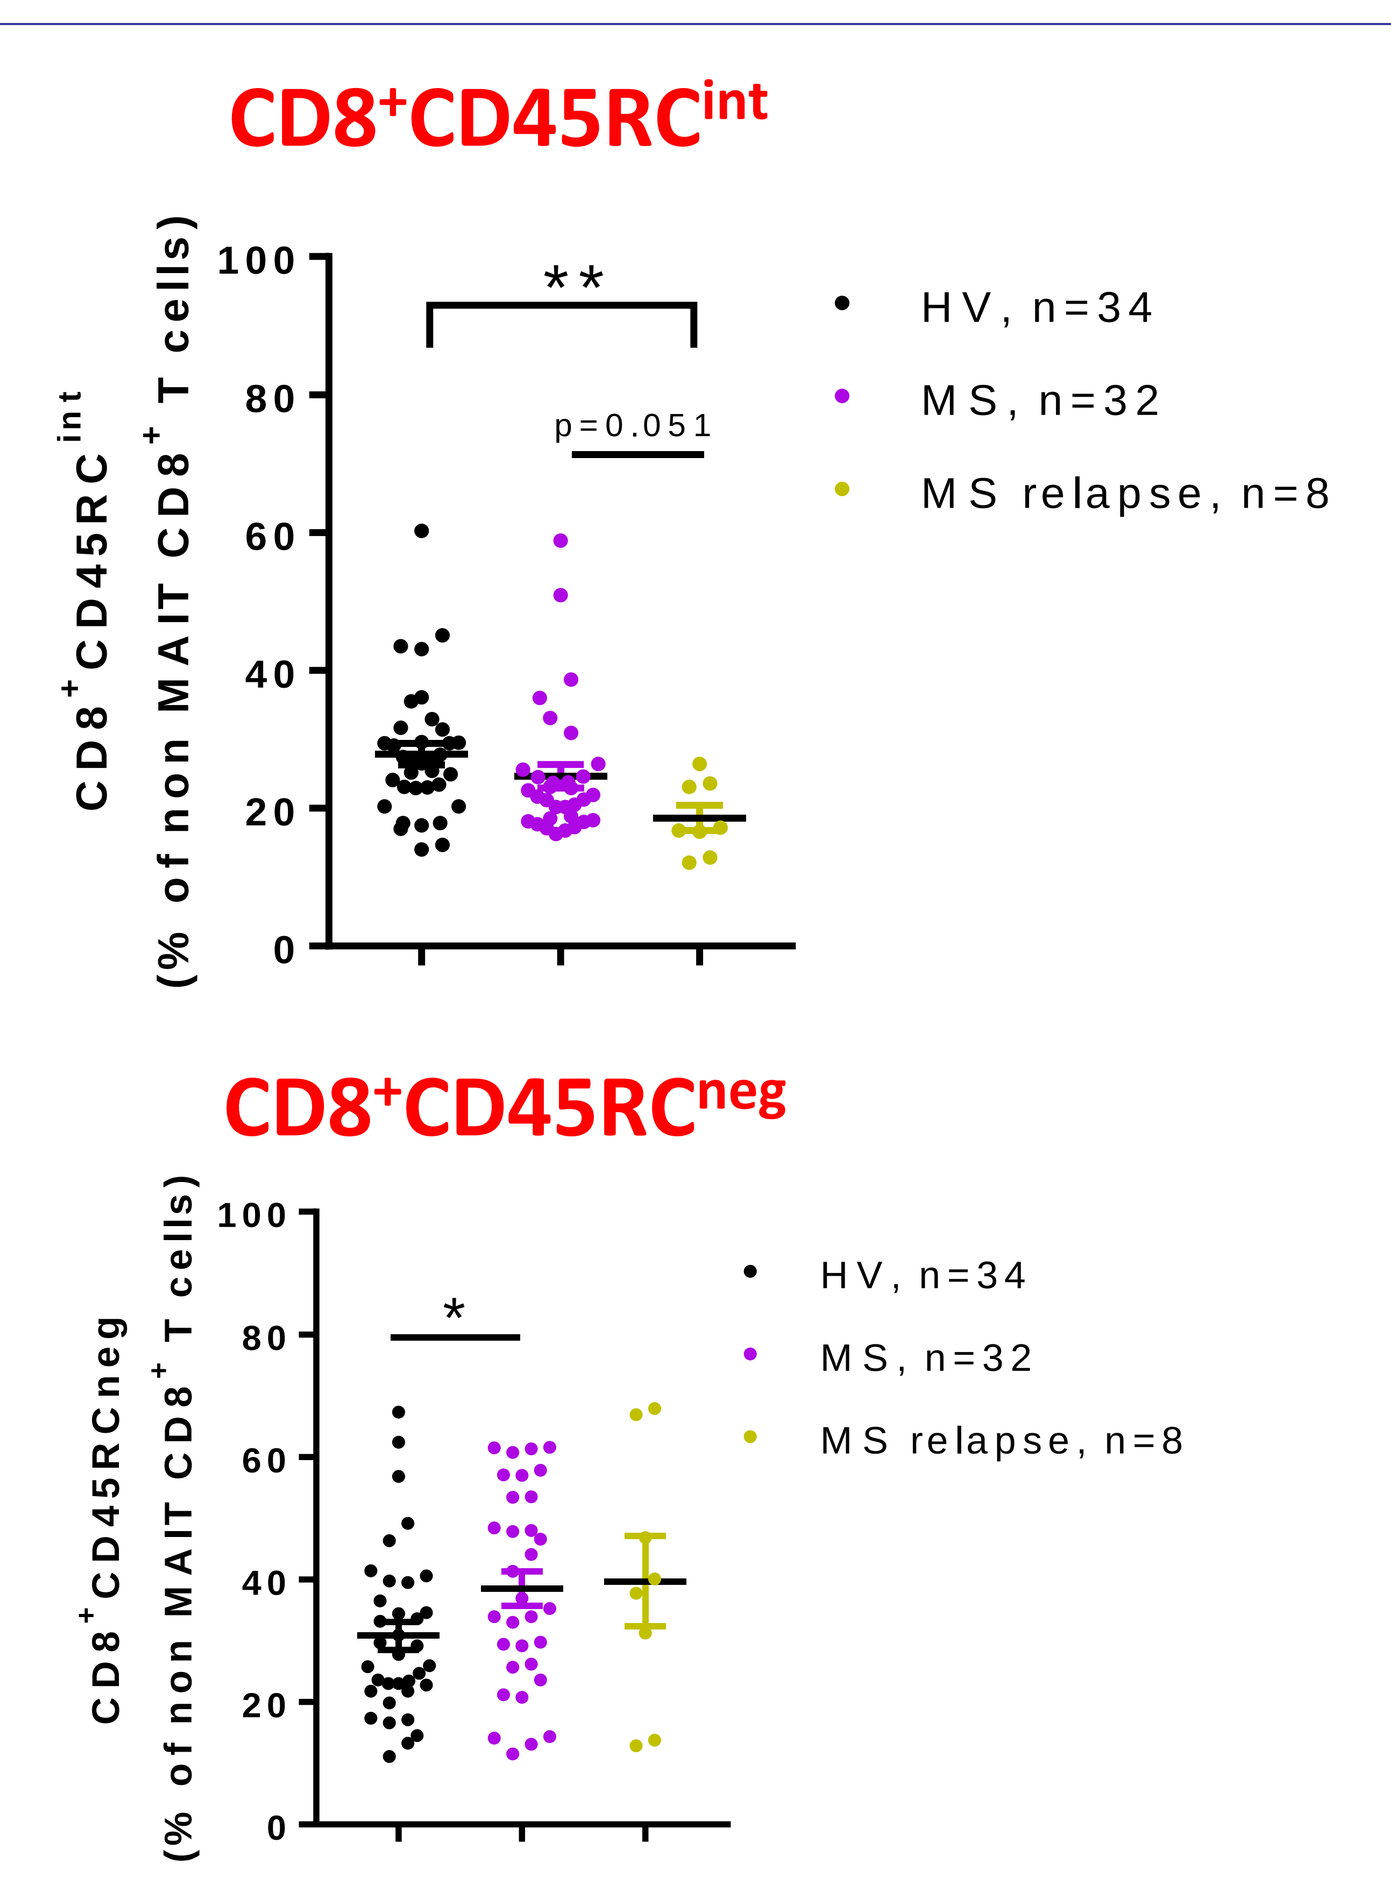
CD8+CD45RC int
**
p=0.051
HV, n=34
MS, n=32
MS relapse, n=8
CD8+CD45RC neg
*

Suppression assay

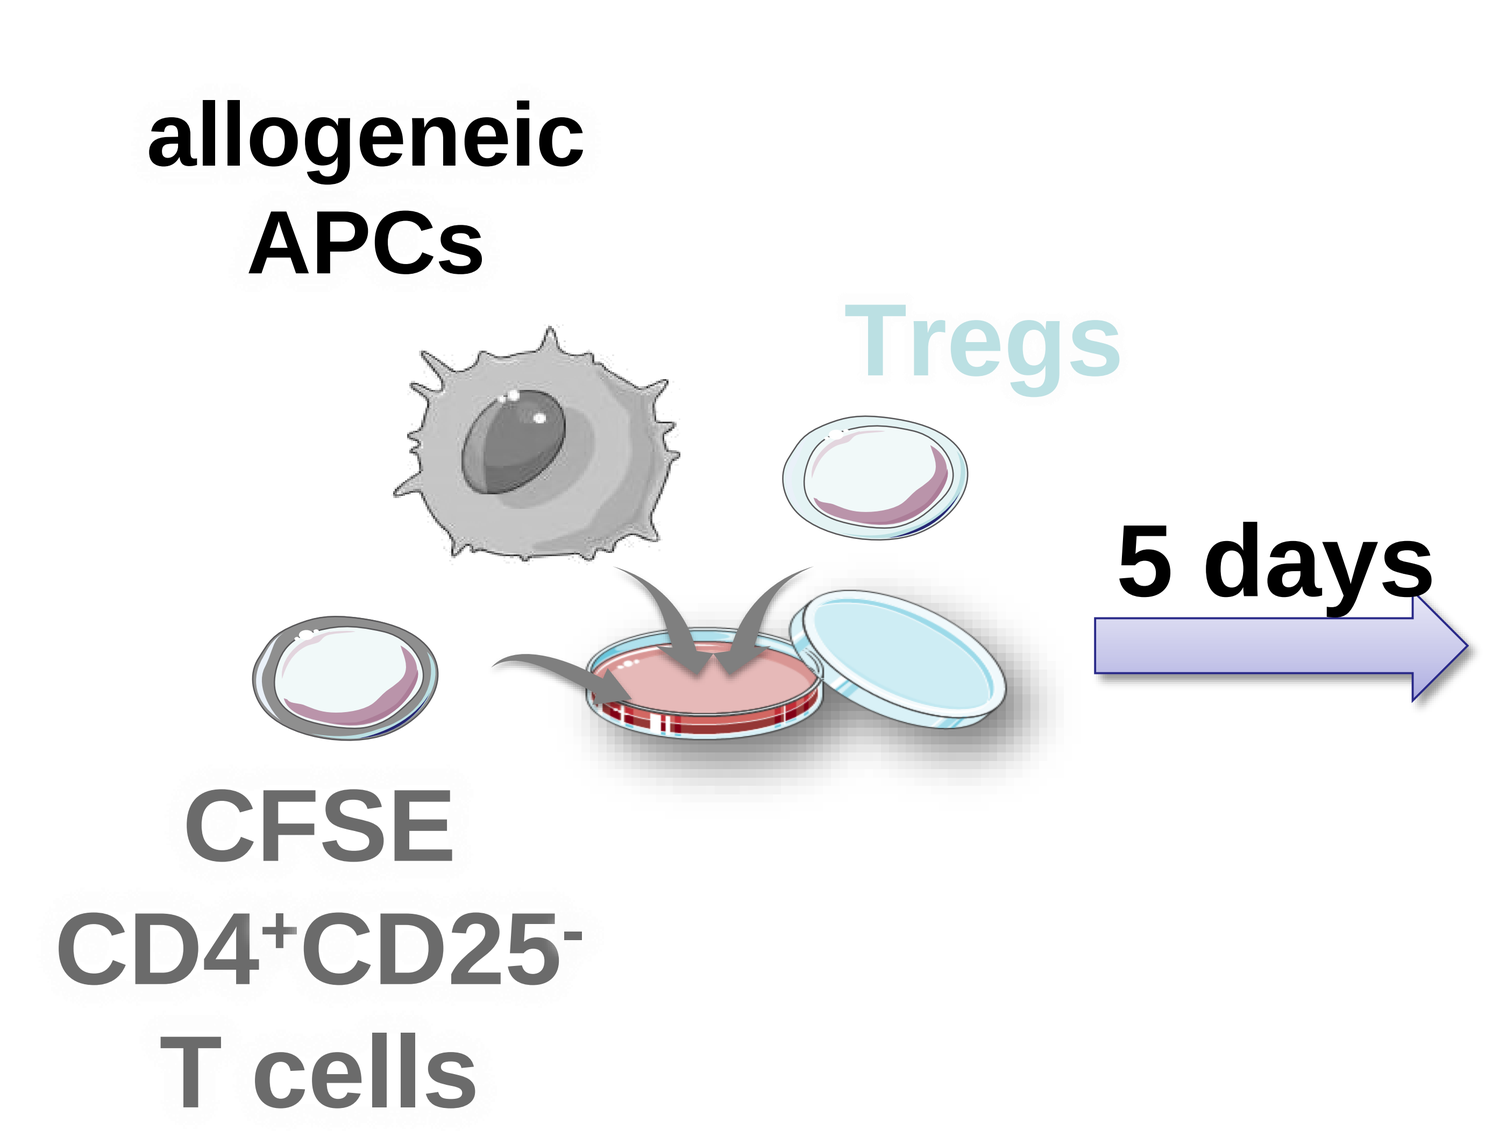
allogeneic APCs
Tregs
5 days
CFSE CD4+CD25- T cells

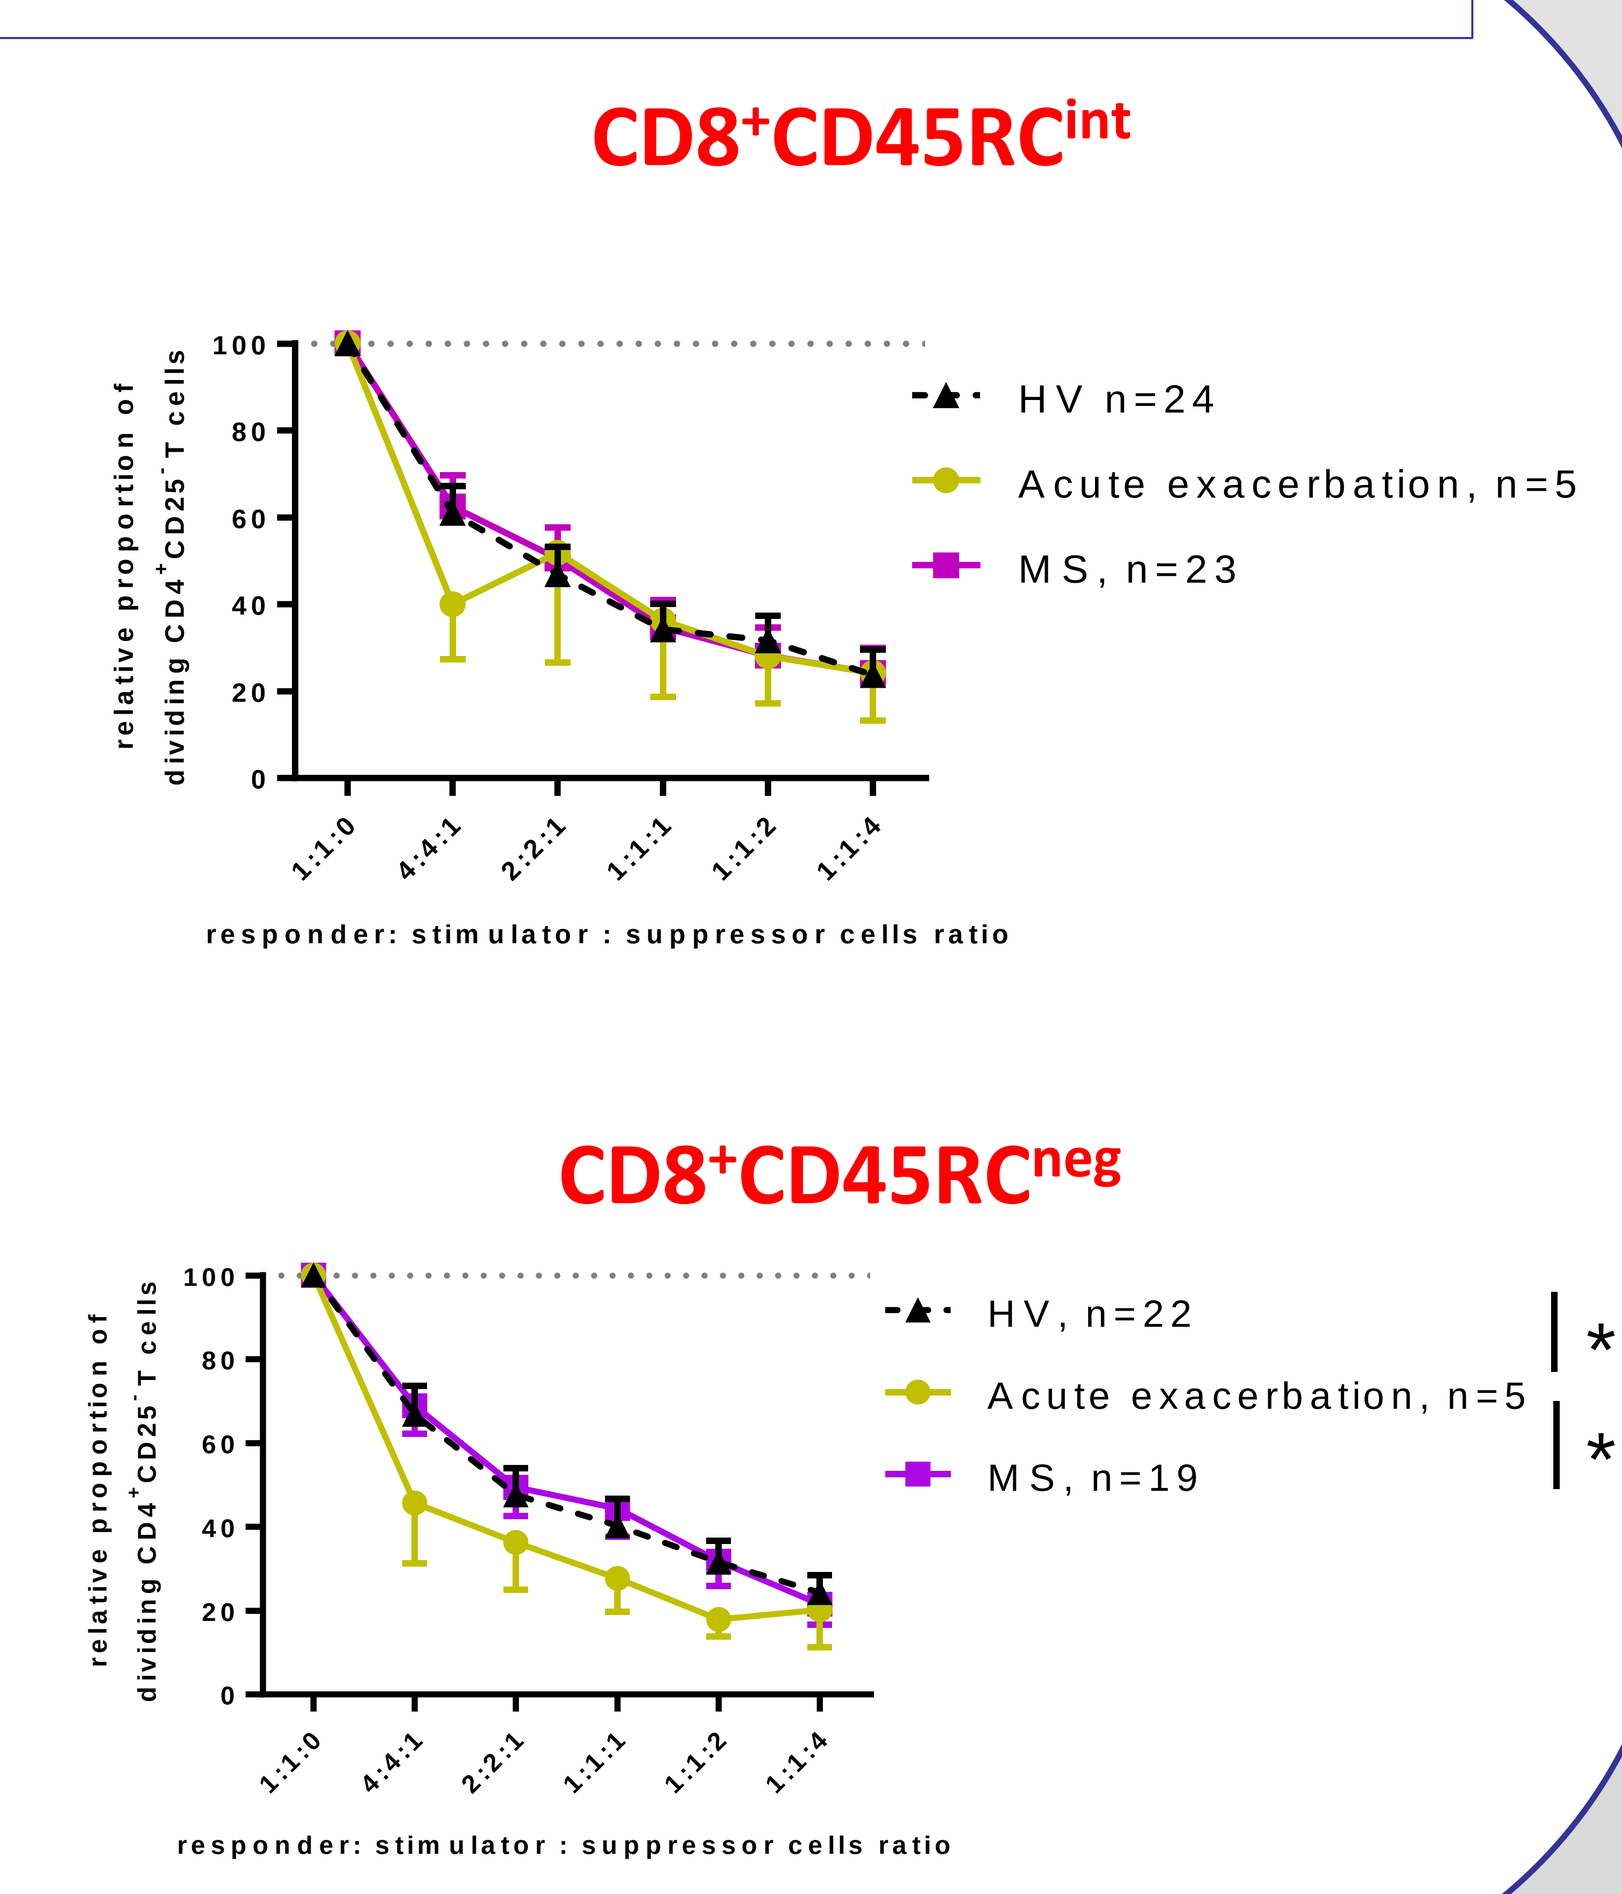
CD8+CD45RC int
HV n=24
Acute exacerbation, n=5
MS, n=23
CD8+CD45RC neg
HV, n=22
Acute exacerbation, n=5
MS, n=19
relative proportion of dividing CD4+CD25- T cells
responder: stimulator : suppressor cells ratio

In severe patients with a higher Multiple Sclerosis Severity Score, CD8+CD45RC^INT Tregs function is impaired

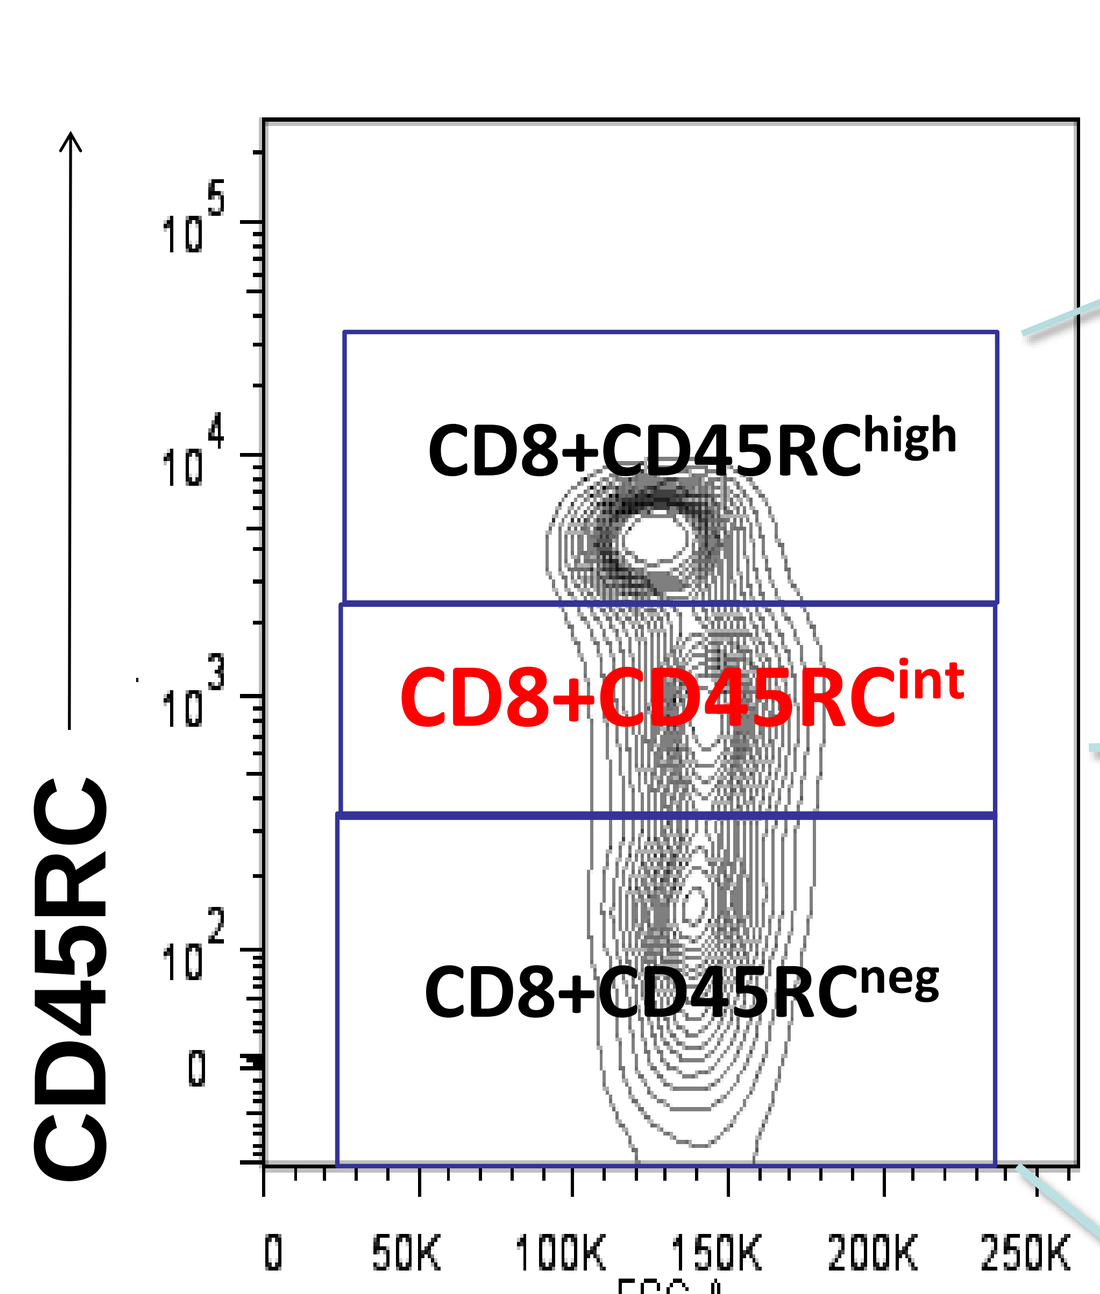
CD45RC
CD8+CD45RChigh
CD8+CD45RCint
CD8+CD45RCneg
FSC-A

Suppression assay

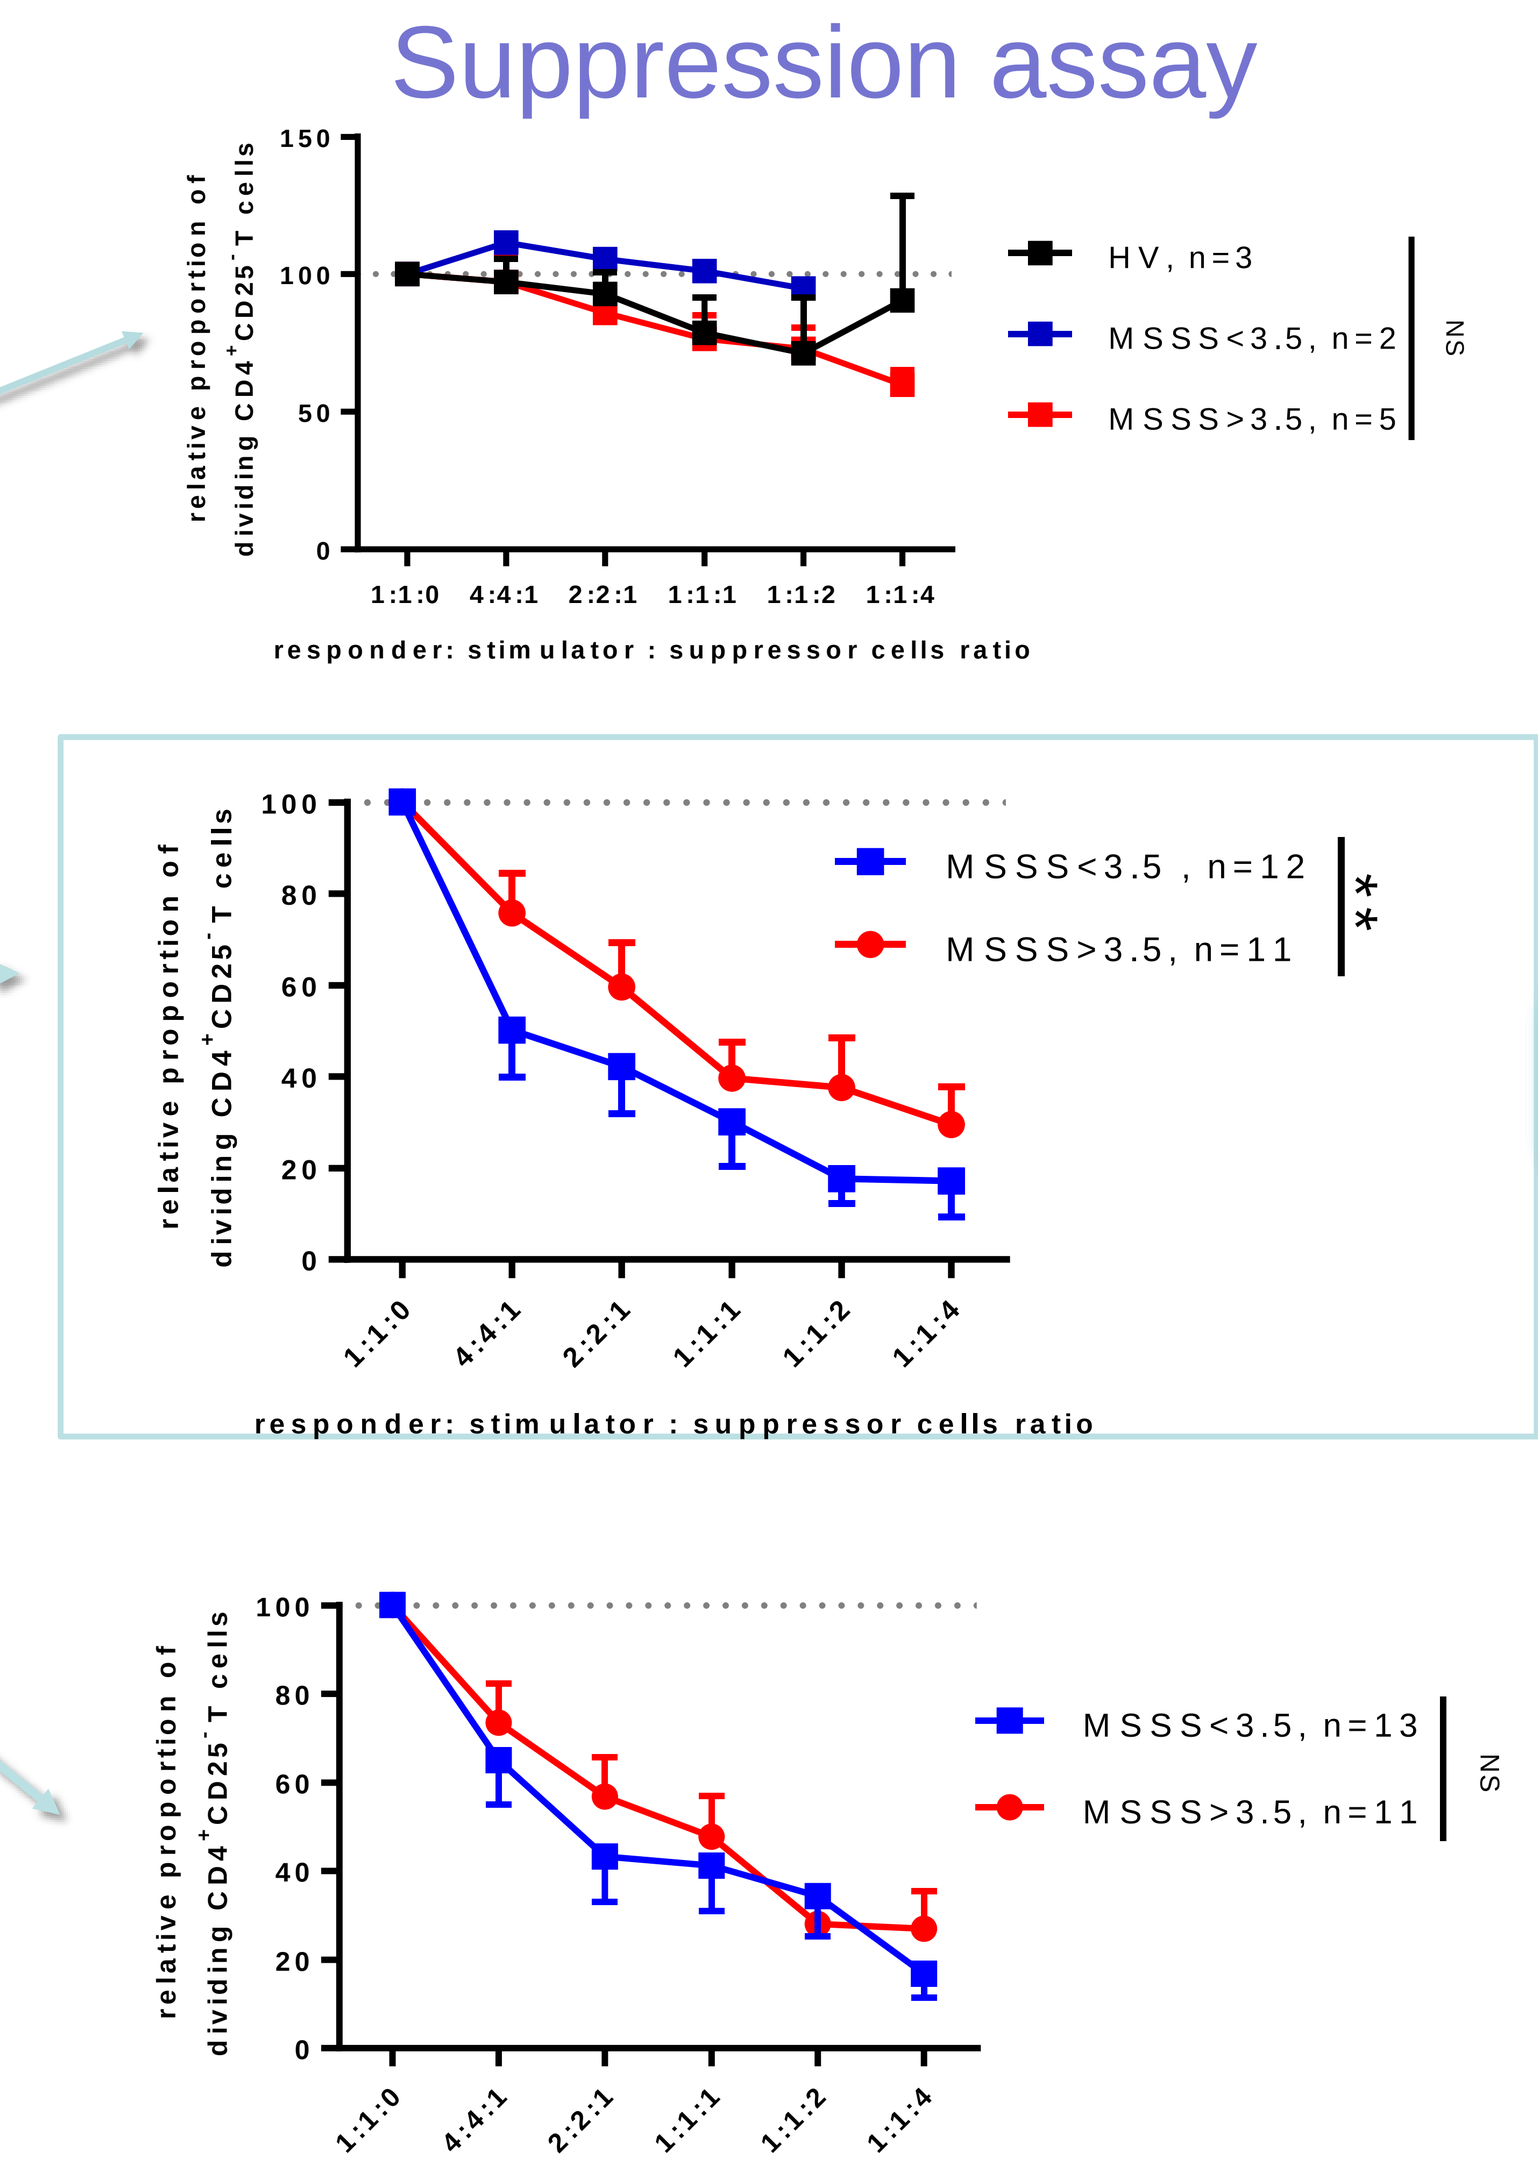
HV, n=3
MSSS<3.5, n=2
MSSS>3.5, n=5
NS
MSSS<3.5 , n=12
MSSS>3.5, n=11
**
MSSS<3.5, n=13
MSSS>3.5, n=11
NS
relative proportion of dividing CD4+CD25- T cells
responder: stimulator : suppressor cells ratio

Transcriptomic Analysis (GSEA)

Regulation of Inflammatory response | **Cytokine-mediated Signaling Pathway**

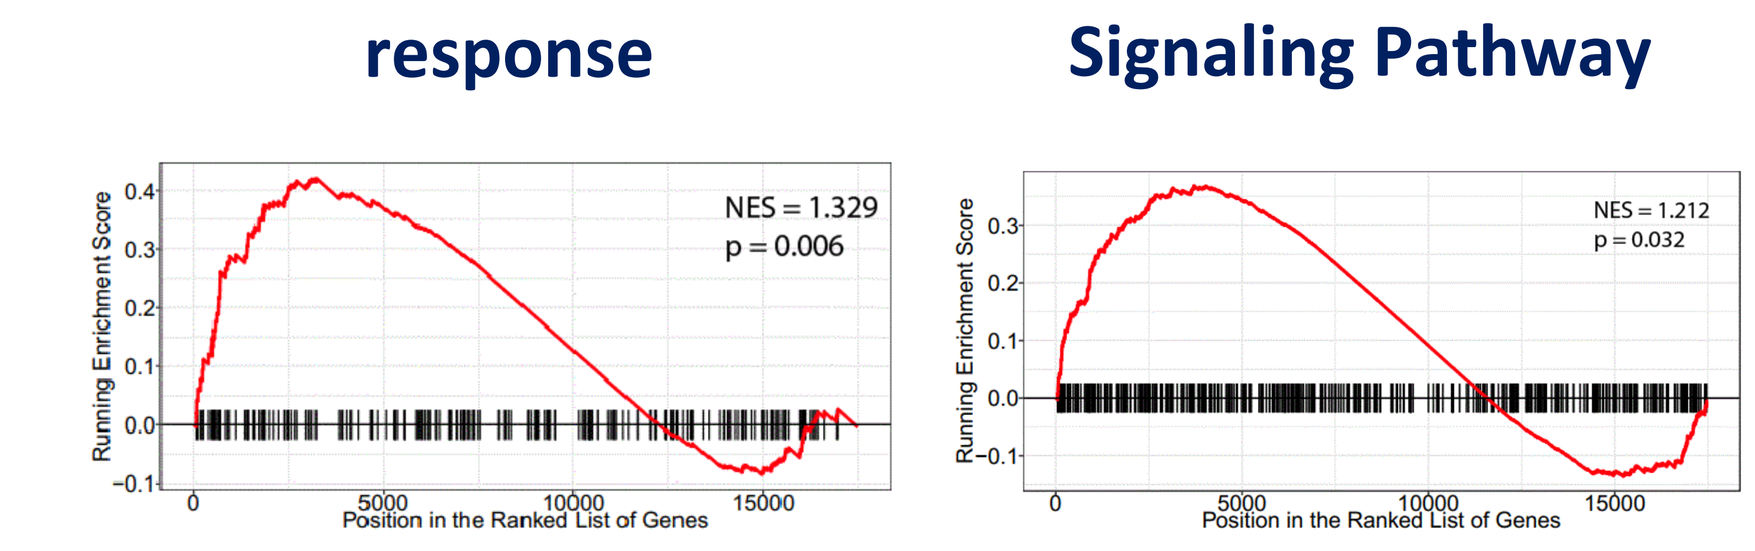
NES = 1.329
p = 0.006
NES = 1.212
p = 0.032
Position in the Ranked List of Genes
Running Enrichment Score

Immune response pathways are overrepresented in CD8+CD45RC^{int} of MSSS<3,5 patients as compared to MSSS>3,5 patients

Flow Cytometry Analysis

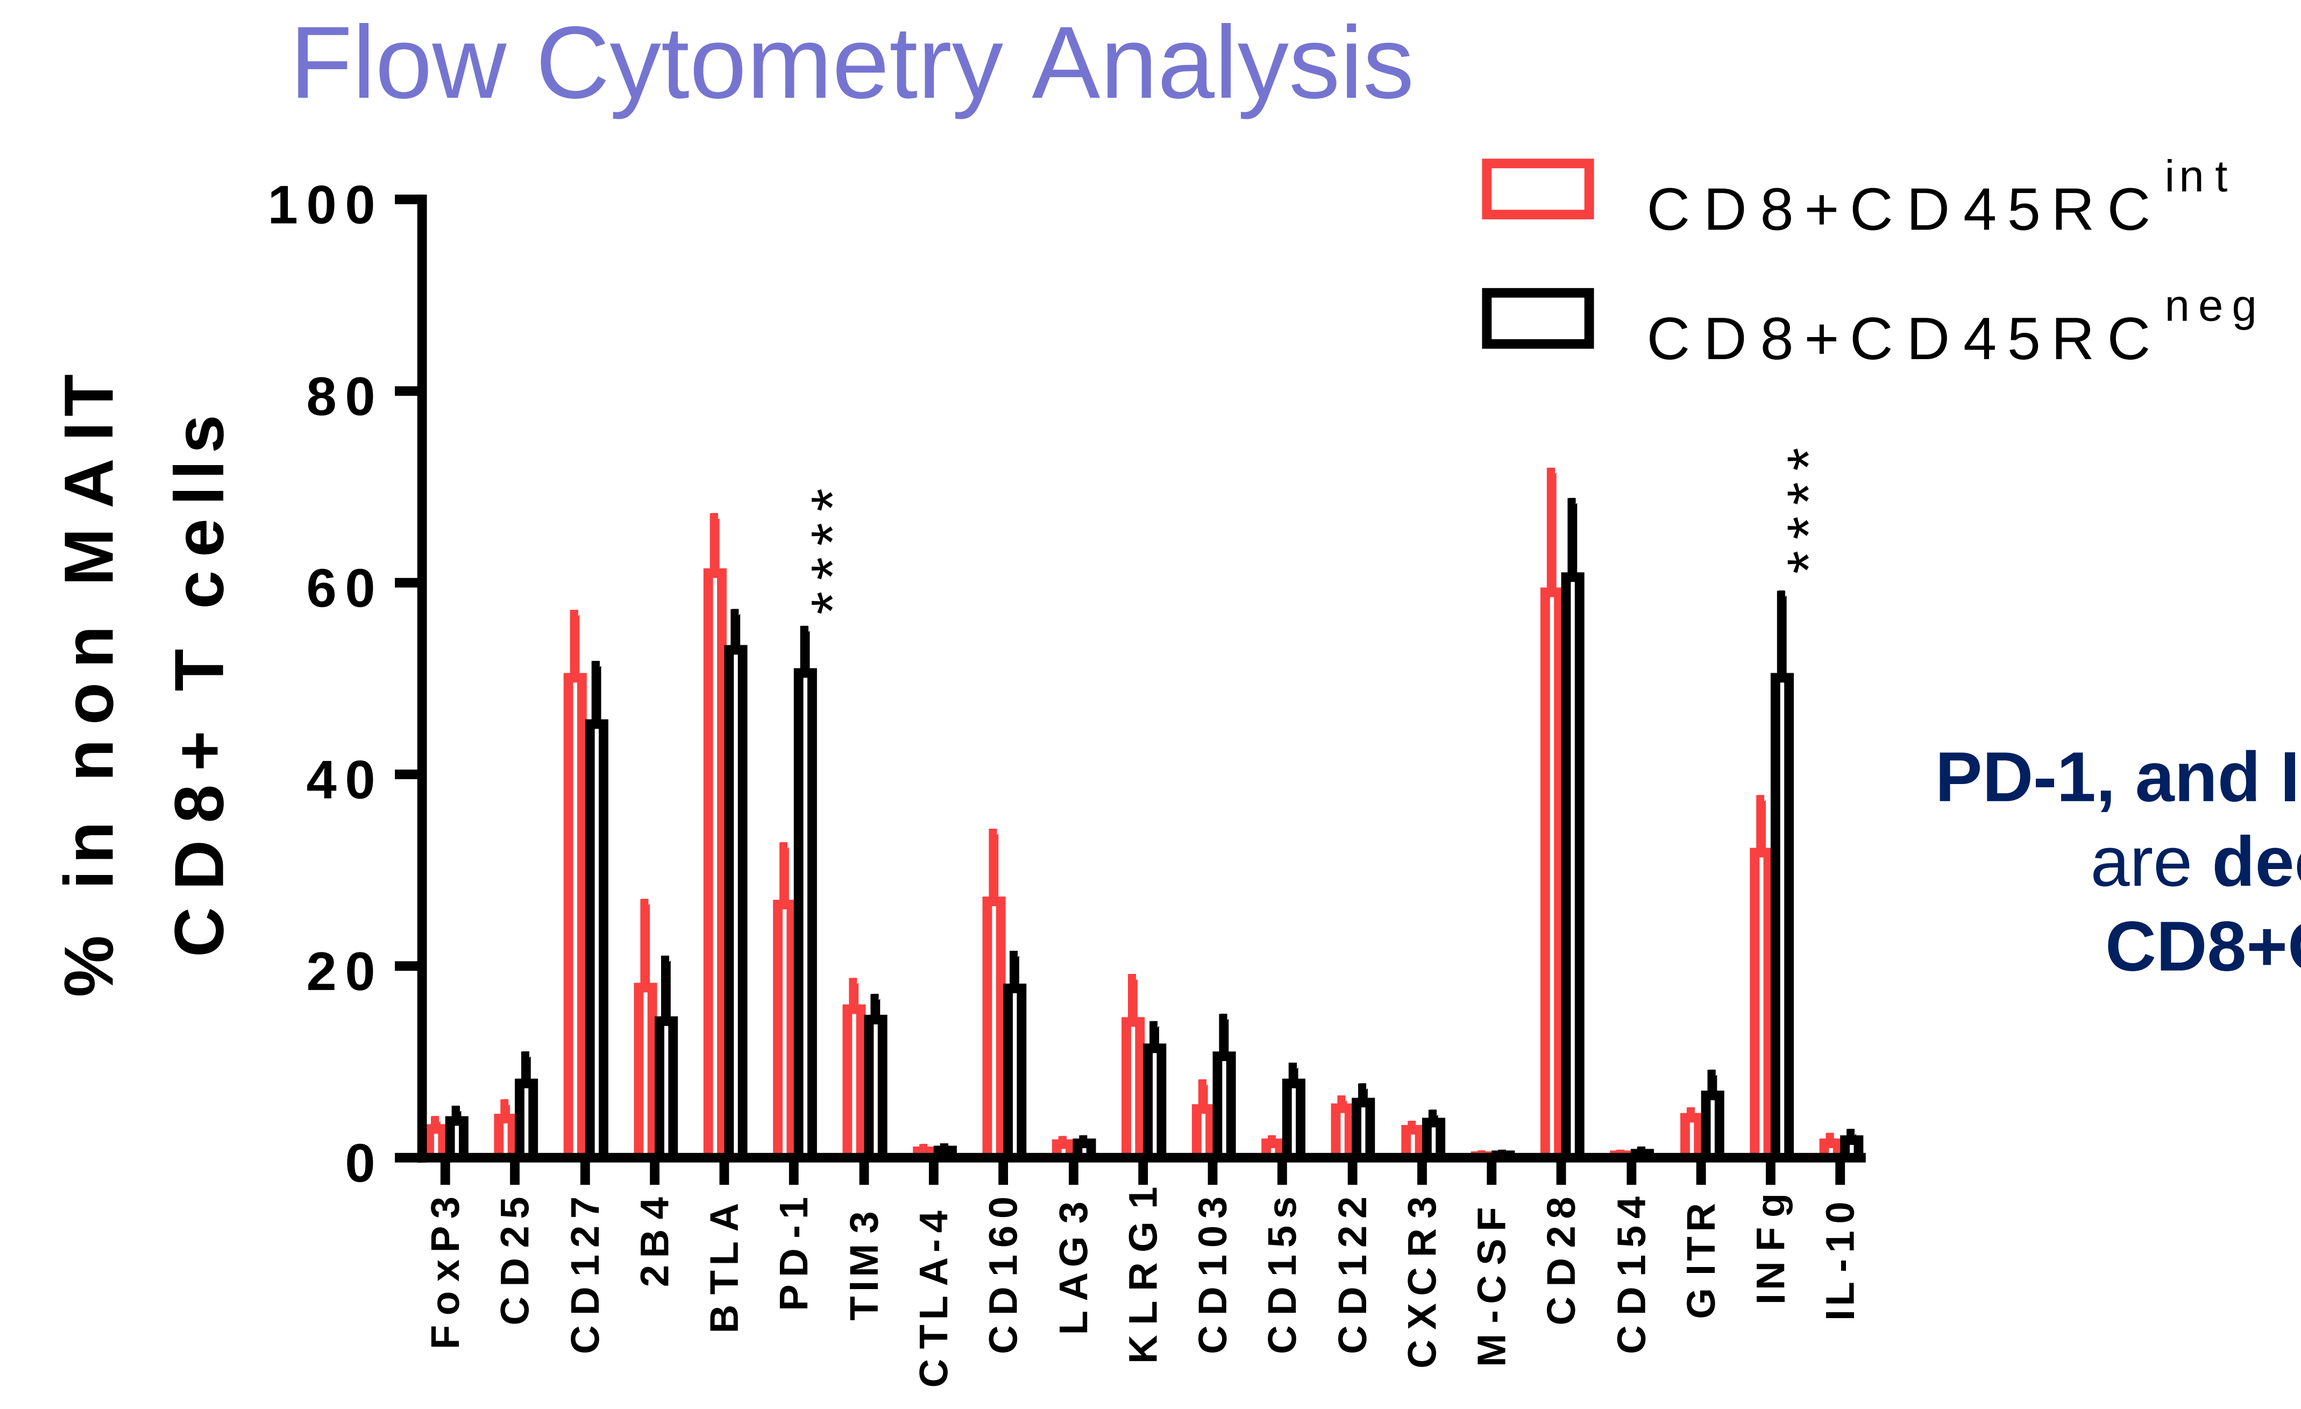
% in non MAIT CD8+ T cells
CD8+CD45RC int
CD8+CD45RC neg

FoxP3
CD25
CD127
2B4
BTLA
PD-1
TIM3
CTLA-4
CD160
LAG3
KLRG1
CD103
CD15s
CD122
CXCR3
M-CSF
CD28
CD154
GITR
INFg
IL-10

PD-1, and IFNg expression are **decreased in CD8+CD45RC^{int}**

Conclusion

For the first time, we demonstrate an **impairment of CD8+CD45RC^{int} Tregs in MS**.

We propose to define two populations of CD8+CD45RC Tregs:

- **CD8+CD45RC^{int} are dysfunctional in severe MS** patients and **less frequent** during exacerbations
- **CD8+CD45RC^{neg} Tregs react properly to inflammation** with enhanced regulatory functions during exacerbations

We suggest CD8$^+$CD45RC$^{int/neg}$ T cells and subsets may be potential therapeutic targets and prognostic tools in MS

Authors have no financial disclosures that would be a potential conflict of interest with this presentation